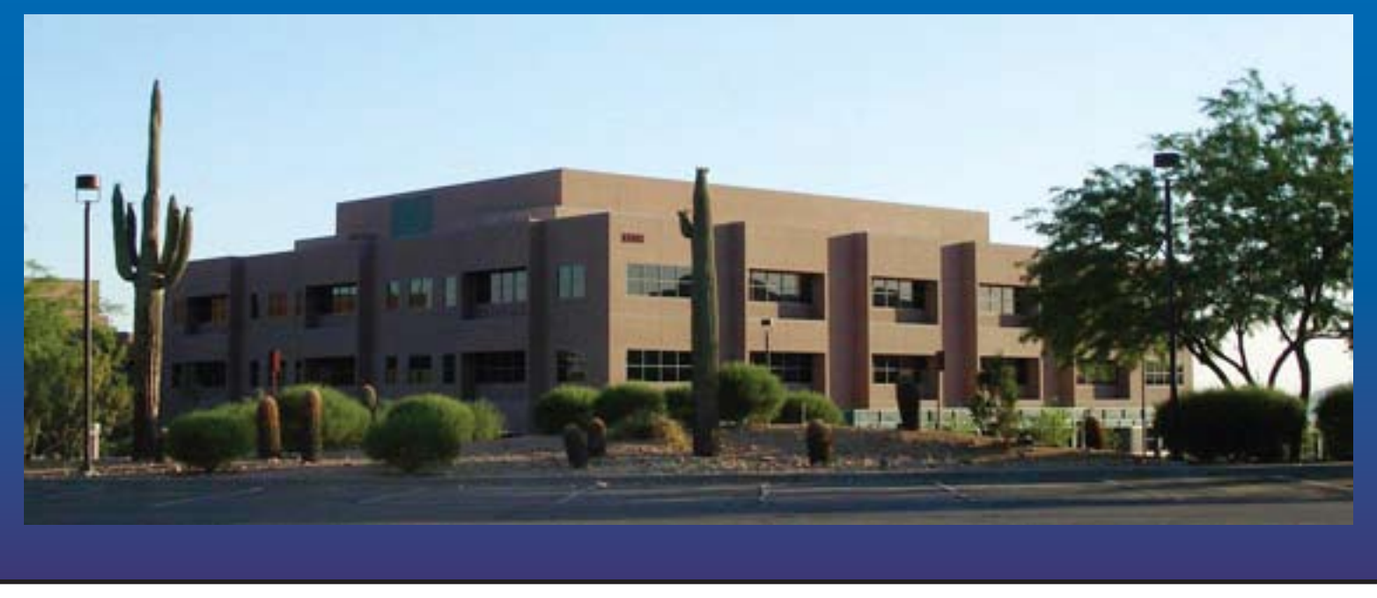

OXIDATIVE MODIFICATION OF AN ANTI-CD20 ANTIBODY IMPARTS AUTOPHILIC BINDING KINETICS

Fan Gao[1], Thomas J. Kindt[1], and Christopher A. Lipinski[2]

[1]InNexus Biotechnology, Scottsdale, AZ 85259; [2]Mayo Clinic Arizona, Scottsdale, AZ 85259

AACR# 1230

Introduction:

Antibody-antibody interactions that occur at the site of antigen binding are the hallmark of "autophilic" binding activity. Homodimerizing antibodies are rarely observed in nature and are typified by the mouse anti-phosphorylcholine antibody (TEPC-15). Autophilic interactions effectively increase the avidity (strength of binding) of the antibody-antigen association that result in measureable kinetic changes. Herein we report a method for controlled-oxidative modification of an anti-CD20 therapeutic antibody that selectively converts kappa light chain tryptophan residues to n-formylkynurenine (NFK) and induces structural changes imparting autophilic properties (referred to as "Wox" = tryptophan oxidation). This Wox anti-CD20 antibody displays typical autophilic binding kinetics, such as increased Bmax and avidity, prolonged dissociation from target, as well as enhancement of effector function. Tryptophan conversion to NFK can be measured by fluorescent emission spectrometry. The precise location of the modification was found to reside in the variable light chain of the antibody as determined by fragmentation analysis. Using computer-modeling analysis, the W to NFK conversion was predicted to increase the solvent-exposed surface area of a variable light region between CDR1-FR2. The empirical surface hydrophobicity was determined by comparing the wild type and Wox antibody uptake of the bis-ANS (1-anilinonaphthalene 8-sulfonate) probe. The Wox antibody had a 17% greater probe uptake than the wild type antibody. The dissociation and association kinetics of DyLight 488 labeled wild type anti-CD20 and Wox were determined using the CD-20 expressing Ramos cell line. The calculated Bmax was ~ 60% higher for the Wox antibody compared to the WT, however this increase may be even greater than the estimated value because of non-saturable binding behavior of Wox. The dissociation of the Wox-CD20 complex was significantly slower than WT with ~40% Wox-CD20 bound to the target even after 5 hours. Potency comparison of WT to Wox (effector dependent and independent) suggests an enhancement for the modified antibody. Thus, Wox anti-CD20 has favorable kinetic attributes that enhance in vitro potency.

Methods:

Detection of Trp Oxidation

Antibody samples were loaded into a 250 μL quartz cuvette for fluorescence measurement using spectrofluorometer FluoMax-4 (Horiba Jobin-Yvon, Irvine, CA). To monitor intrinsic protein fluorescence, emission scanning was performed with excitation wavelength centered at 295 nm and 320 nm separately.

Measurement of Antibody Hydrophobicity

In order to monitor changes in protein hydrophobicity, antibody samples were assayed with bis-ANS (Invitrogen, Carlsbad, CA), and the resulting fluorescence was monitored in emission spectra with excitation wavelength at 384 nm. In brief, equal amount of Bis-ANS was added to the diluted antibody samples, and incubated for 15 min. before measurement. The fluorescence intensities collected at 500 nm were used for analysis. The data was analyzed in Excel 2007 and fitted by linear regression model.

Study of Antibody Binding Kinetics

Association and dissociation kinetics of DyLight-448 labeled mAbs to CD20-bearing Ramos cells were measured by determination of the amount of CD20-bound labeled antibody over time. Dissociation of bound antibody was triggered by adding a saturating concentration of non-labeled antibody (1 μM) to cells that have been pre-incubated with labeled antibody (10 nM) for 1 hour. Non-specific binding was determined by incubating cells with labeled antibody and saturating amount of non-labeled antibody simultaneously. Fluorescence intensity of bound labeled antibodies was measured from Synergy2 Microplate Reader (Biotek, Winooski, VT) with Ex/Em wavelength centered at 485/528. Binding data were analyzed and plotted using Prism 5 (GraphPad Software, La Jolla, CA).

Antibody binding kinetics to CD20 mimic epitope was measured using BIAcore T100. A biotinylated CD20 peptidomimetic was immobilized on an avidin-coated chip. Antibodies were injected at a flow rate of 30μL/min at 25 ˚C. Association phase is measured by calculating the reflective angle of an incident wavelength of light off the under-surface of the chip to create a sensorgram. Following the binding phase, the surface is regenerated at the end of each cycle by a quick injection of glycine buffer.

Study of in-vitro potency

Growth Inhibition. Ramos cells were seeded into 96-well plates and treated with antibody for 24 hours. The ATP-based luminescent CellTiter-Glo Reagent (Promega, Madison, WI) was subsequently added and incubated with cells for 1 hour. Luminescence was read from Synergy2 Microplate Reader.

Antibody-dependent cellular cytotoxicity. CD56+ natural killer (NK) cells were purified by immunomagnetic separation (Miltenyi Biotec, Auburn, CA) from normal human peripheral blood mononuclear cell (PBMC) samples (AllCells, Emeryville, CA); effector cell purity (CD56+/CD16+) was confirmed by flow cytometry. NK cells were added to Ramos cells 1:6 (effector:target) in the presence or absence of drug (10 μg/mL) and ATP content was assessed as described above after 24 hours incubation.

Complement-dependent cytotoxicity. Human peripheral blood lymphocyte (PBL) samples (2.5 x 105 cells/well, 0.5 mL/well) were seeded into a 24-well format and treated with 5% (v/v) rabbit HLA-ABC complement sera for 30 minutes, followed by addition of drug (10 μg/mL) for 2 hours. Depletion of CD19+ cells was assessed by flow cytometry (BD FACSCanto II). After identifying the whole lymphocyte population (FSC, SSC gating), the percentage of CD19+ cells was determined from this subset.

Fragmentation of Intact Antibody

Fc and $F(ab)'_2$ domains were generated by papain or pepsin digestion, followed by protein A and protein L affinity purification. Heavy and light chain fragments were prepared by reduction and alkylation, followed by purification using size-exclusion chromatography in acidic condition.

Molecular Modeling

Computer simulation was performed using ICM software (Molsoft, La Jolla, CA). Modeling of intact antibody structure was conducted using Homology modeling routine. Structural perturbation was simulated using biased probability Monte Carlo routine.

Fig. 1. Autophilic properties to an anti-CD20 mAb imparted by oxidative modifications of Trp -- "Wox" mAb

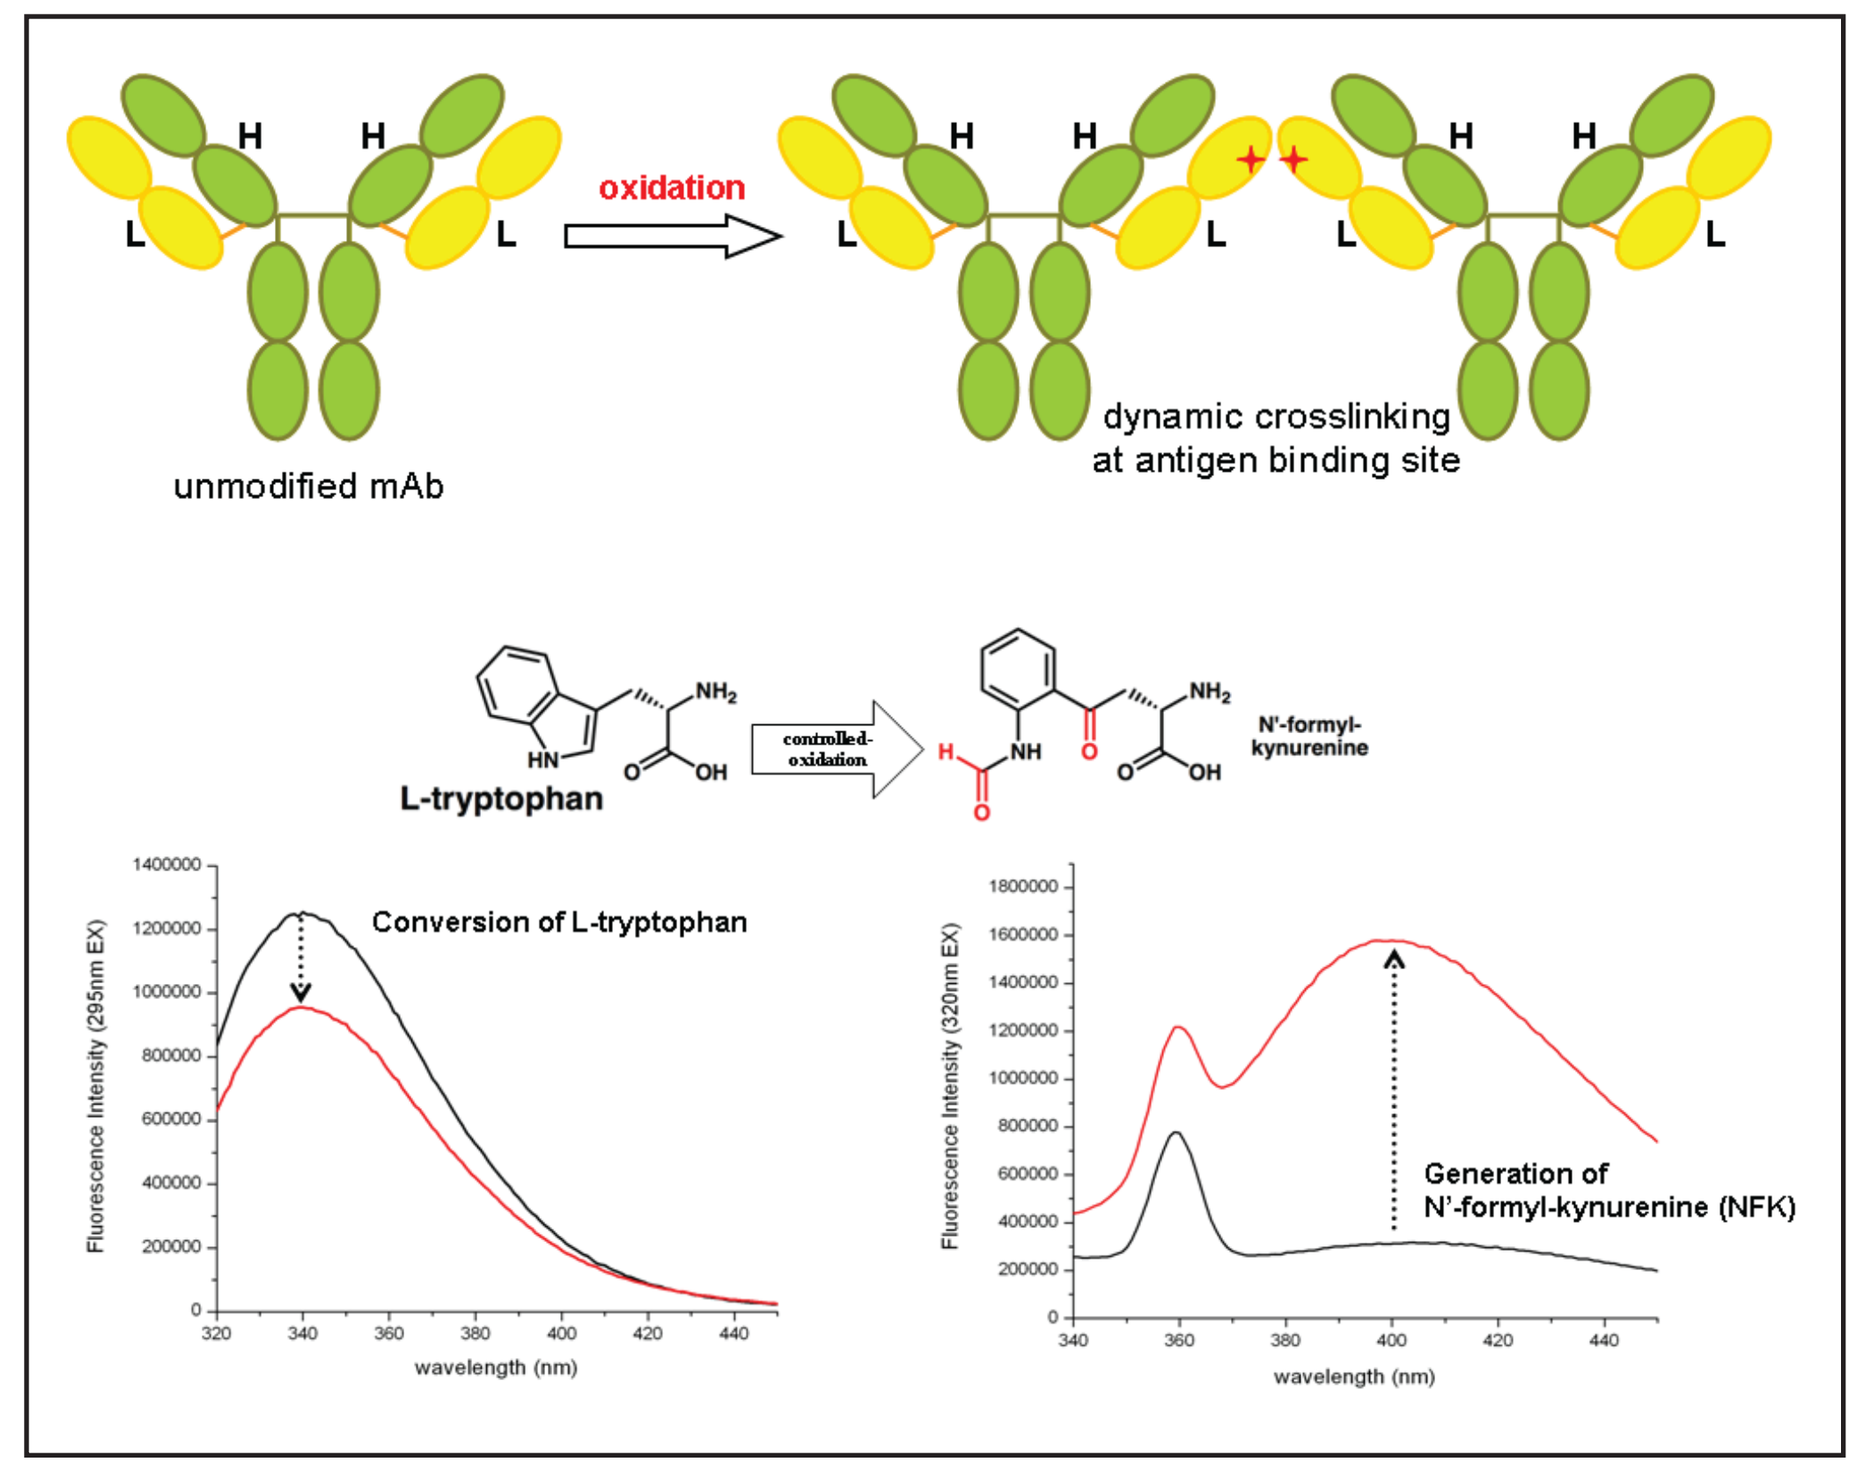

H
L
oxidation
unmodified mAb
dynamic crosslinking
at antigen binding site
L-tryptophan
controlled-oxidation
N'-formyl-kynurenine
Conversion of L-tryptophan
Generation of
N'-formyl-kynurenine (NFK)
Fluorescence Intensity (295nm EX)
Fluorescence Intensity (320nm EX)
wavelength (nm)

Upper panels: The scheme of imparting "autophilic" binding to mAb by re-surfacing the antibody using controlled oxidation method.
Lower panels: Oxidation specific to Trp in the antibody was detected by spectro-fluorometry (reduction of Trp fluorescence and generation of fluorescence from its oxidized form represented as NFK).

Fig. 2. Increased solvent exposure of hydrophobic patches of "Wox" mAb

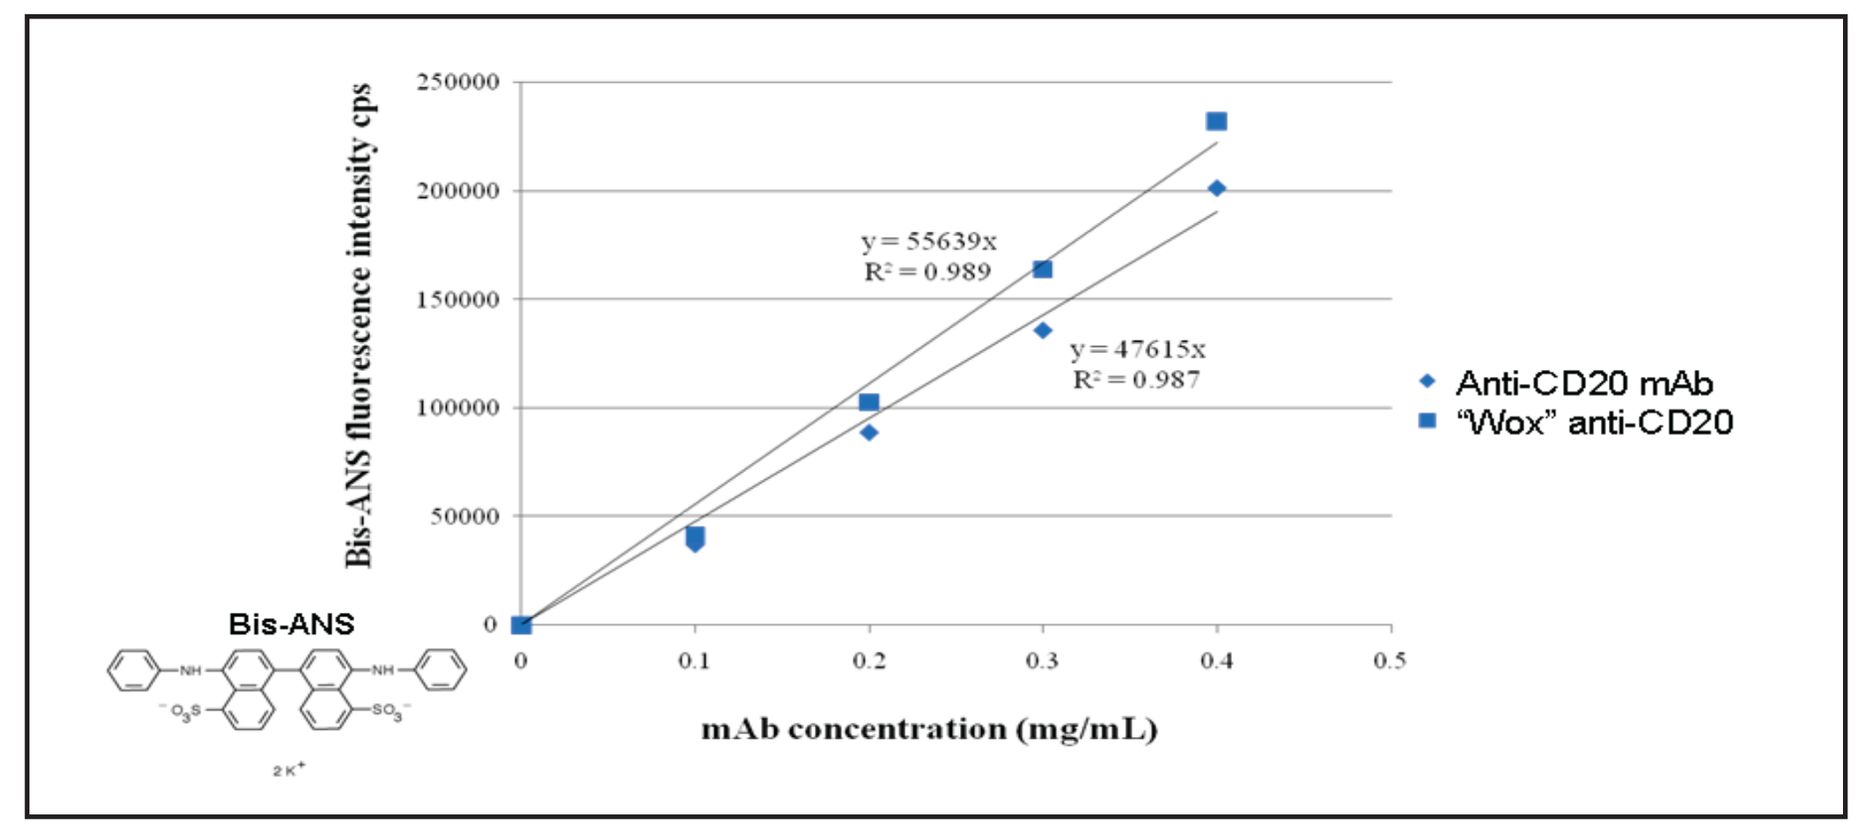

Bis-ANS fluorescence intensity cps
y = 55639x
R² = 0.989
y = 47615x
R² = 0.987
Anti-CD20 mAb
"Wox" anti-CD20
Bis-ANS
mAb concentration (mg/mL)

Exposure of hydrophobic patches was determined by measurement of fluorescence intensity of hydrophobic fluorescent probe bis-ANS (4,4'-dianilino-1,1'-binaphyl-5,5'-disulfonic acid, dipotassium salt). In comparison with WT, "Wox" showed ~17% increase in solvent exposure.

Fig. 3. Changed binding kinetics of "Wox" mAb to CD20 antigen

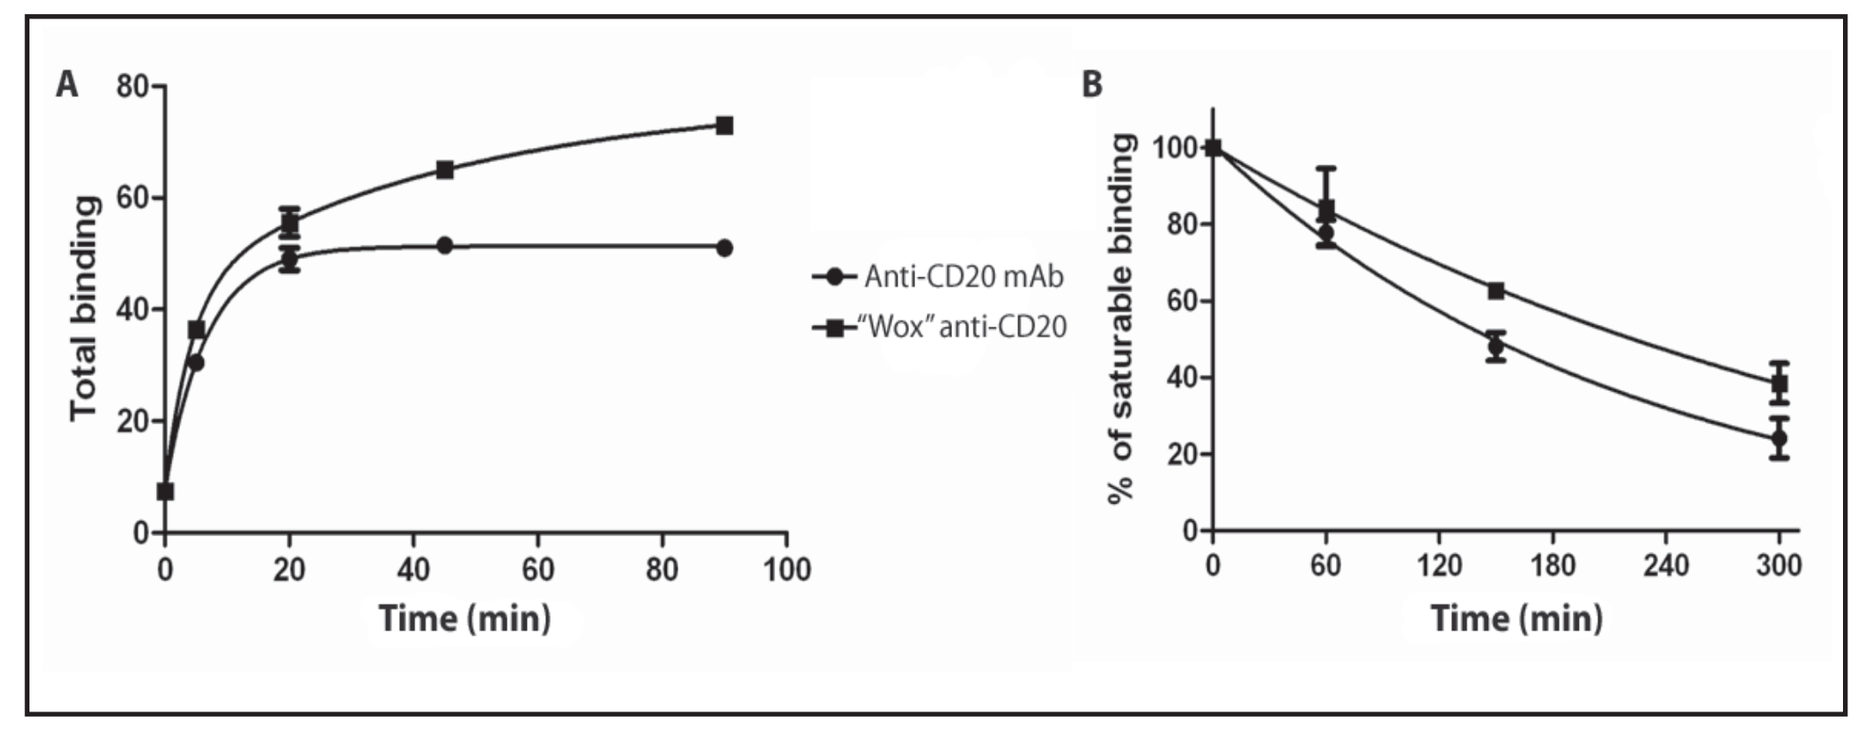

A
B
Total binding
% of saturable binding
Time (min)
Anti-CD20 mAb
"Wox" anti-CD20

Comparison of association kinetics (**Panel A**) and dissociation kinetics (**Panel B**) of non-oxidized and oxidized anti-CD20 using the CD20 expressing Ramos cell line. "Wox" mAb displayed non-saturable binding kinetics with increased Bmax (~60% higher). Association rate *kon* for "Wox" increased ~20% for the initial binding phase (first 20 min.). The dissociation of "Wox" from target was significantly slower with dissociation rate *koff* decreased by ~2 fold.

Fig. 4. Increased potency observed for "Wox" mAb

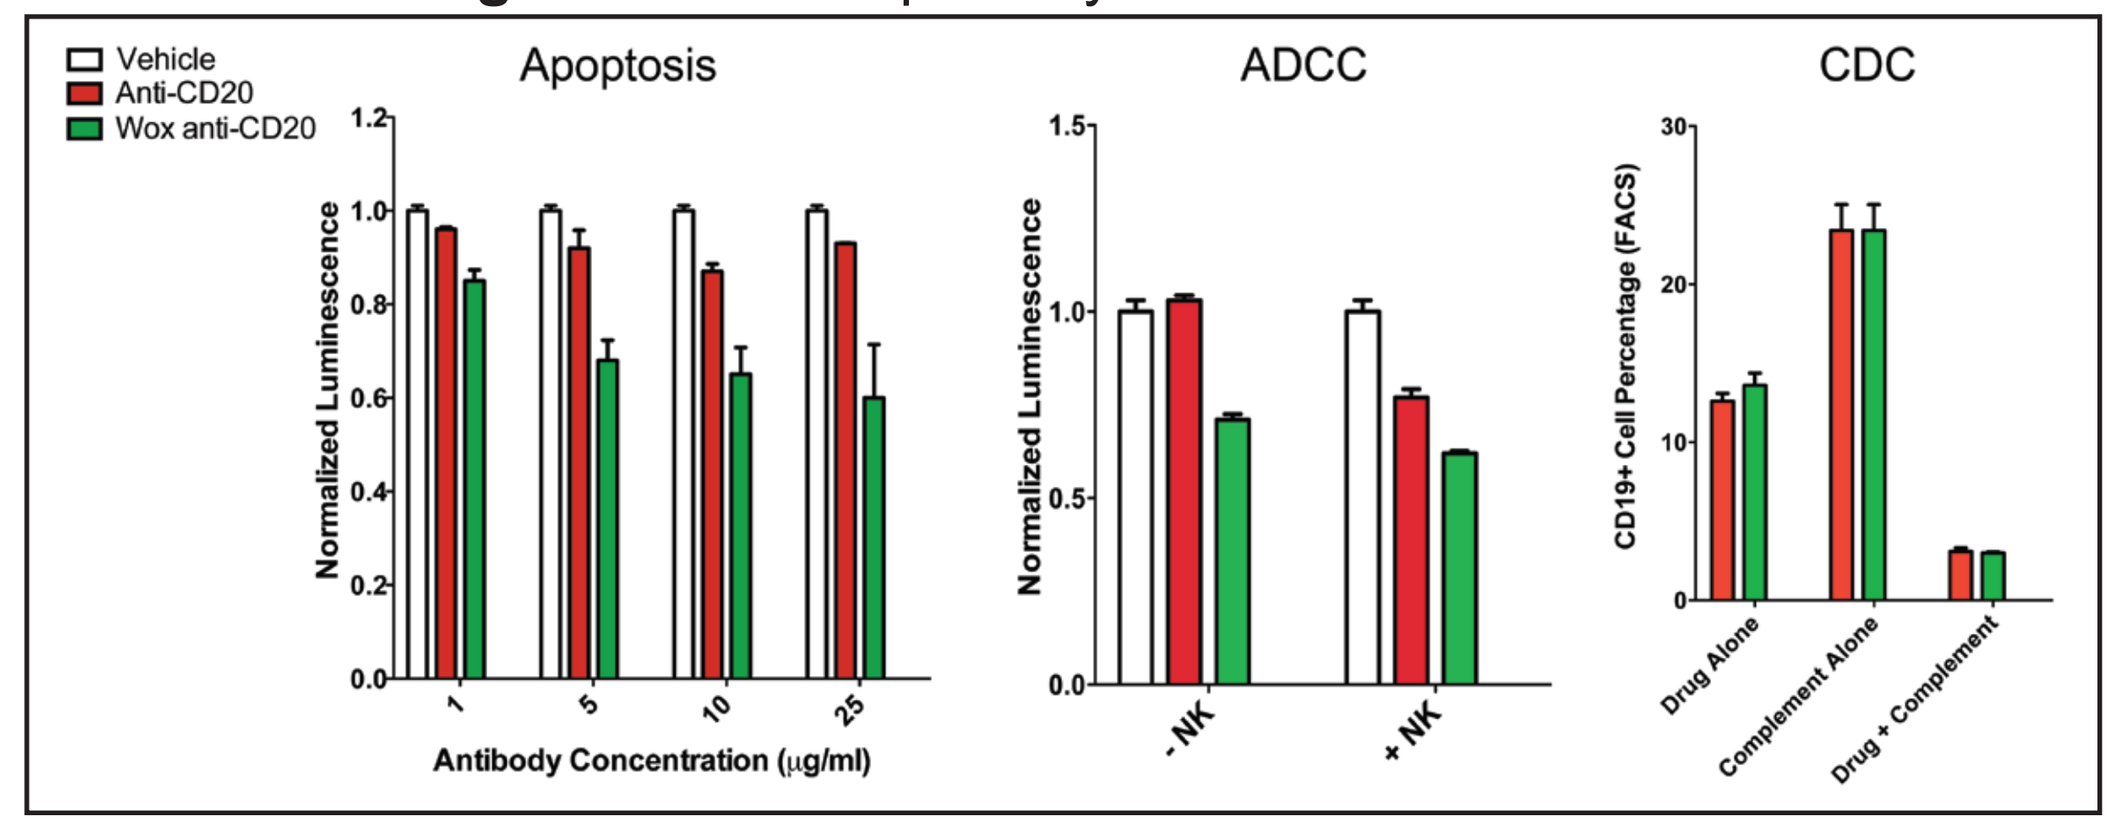

Vehicle
Anti-CD20
Wox anti-CD20
Apoptosis
ADCC
CDC
Normalized Luminescence
Antibody Concentration (μg/ml)
- NK
+ NK
CD19+ Cell Percentage (FACS)
Drug Alone
Complement Alone
Drug + Complement

Apoptosis (Panel A): Ramos cells treated with Wox anti-CD20 had increase in the relative proportion of apoptotic cells when compared to vehicle or anti-CD20. **ADCC (Panel B)**: Ramos cell were pre-incubated for 1 hour with antibody (10 μg/ml) followed by addition of NK cells. Fluorescent viability staining shows that Wox anti-CD20 mediates a greater cell kill than comparator groups. **CDC (Panel C)**: Human PBMCs were pretreated with rabbit complement for 20 minutes followed by the addition of either Wox or unmodified anti-CD20 antibody. Each antibody induced cytotoxicity to a similar extent in the presence of complement. (*p<0.05)

Fig. 5. Oxidation of $F(ab)'_2$ domain responsible for autophilic properties

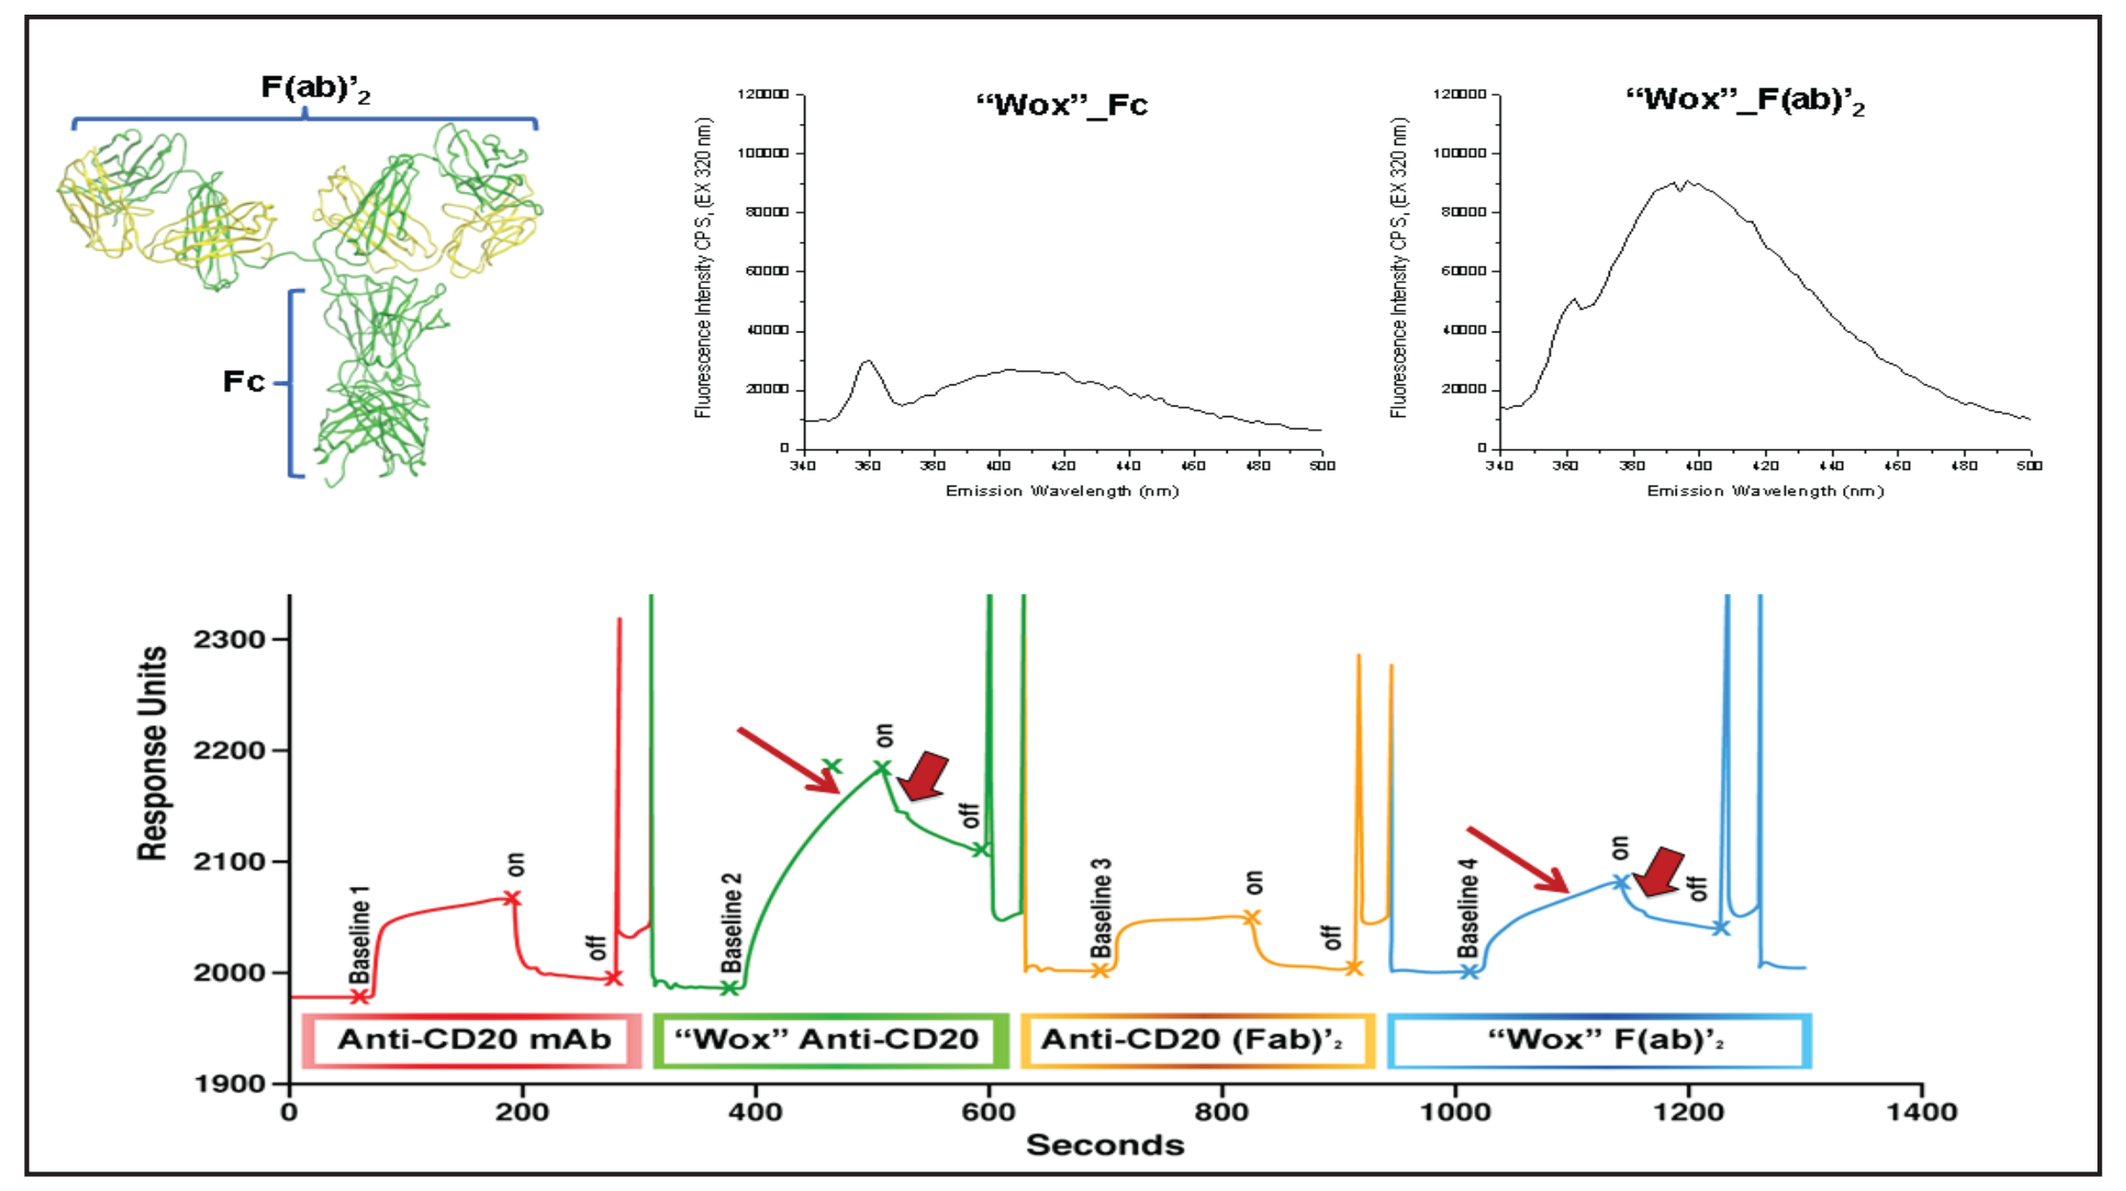

F(ab)'₂
Fc
"Wox"_Fc
"Wox"_F(ab)'₂
Emission Wavelength (nm)
Response Units
Seconds
Baseline 1
Baseline 2
Baseline 3
Baseline 4
on
off
Anti-CD20 mAb
"Wox" Anti-CD20
Anti-CD20 (Fab)'₂
"Wox" F(ab)'₂

Upper panels: Fluorescence emission spectra of "Wox" Fc and $F(ab)'_2$ domains. Trp(s) in $F(ab)'_2$ has been oxidized.
Lower panels: Biacore binding kinetics of non-oxidized and oxidized intact antibody and antibody fragments to CD20 epitope. Autophilic properties were conserved in $F(ab)'_2$ domain.

Fig. 6. Oxidation of Trp in light chain revealed by protein fragmentation and fluorescence measurement

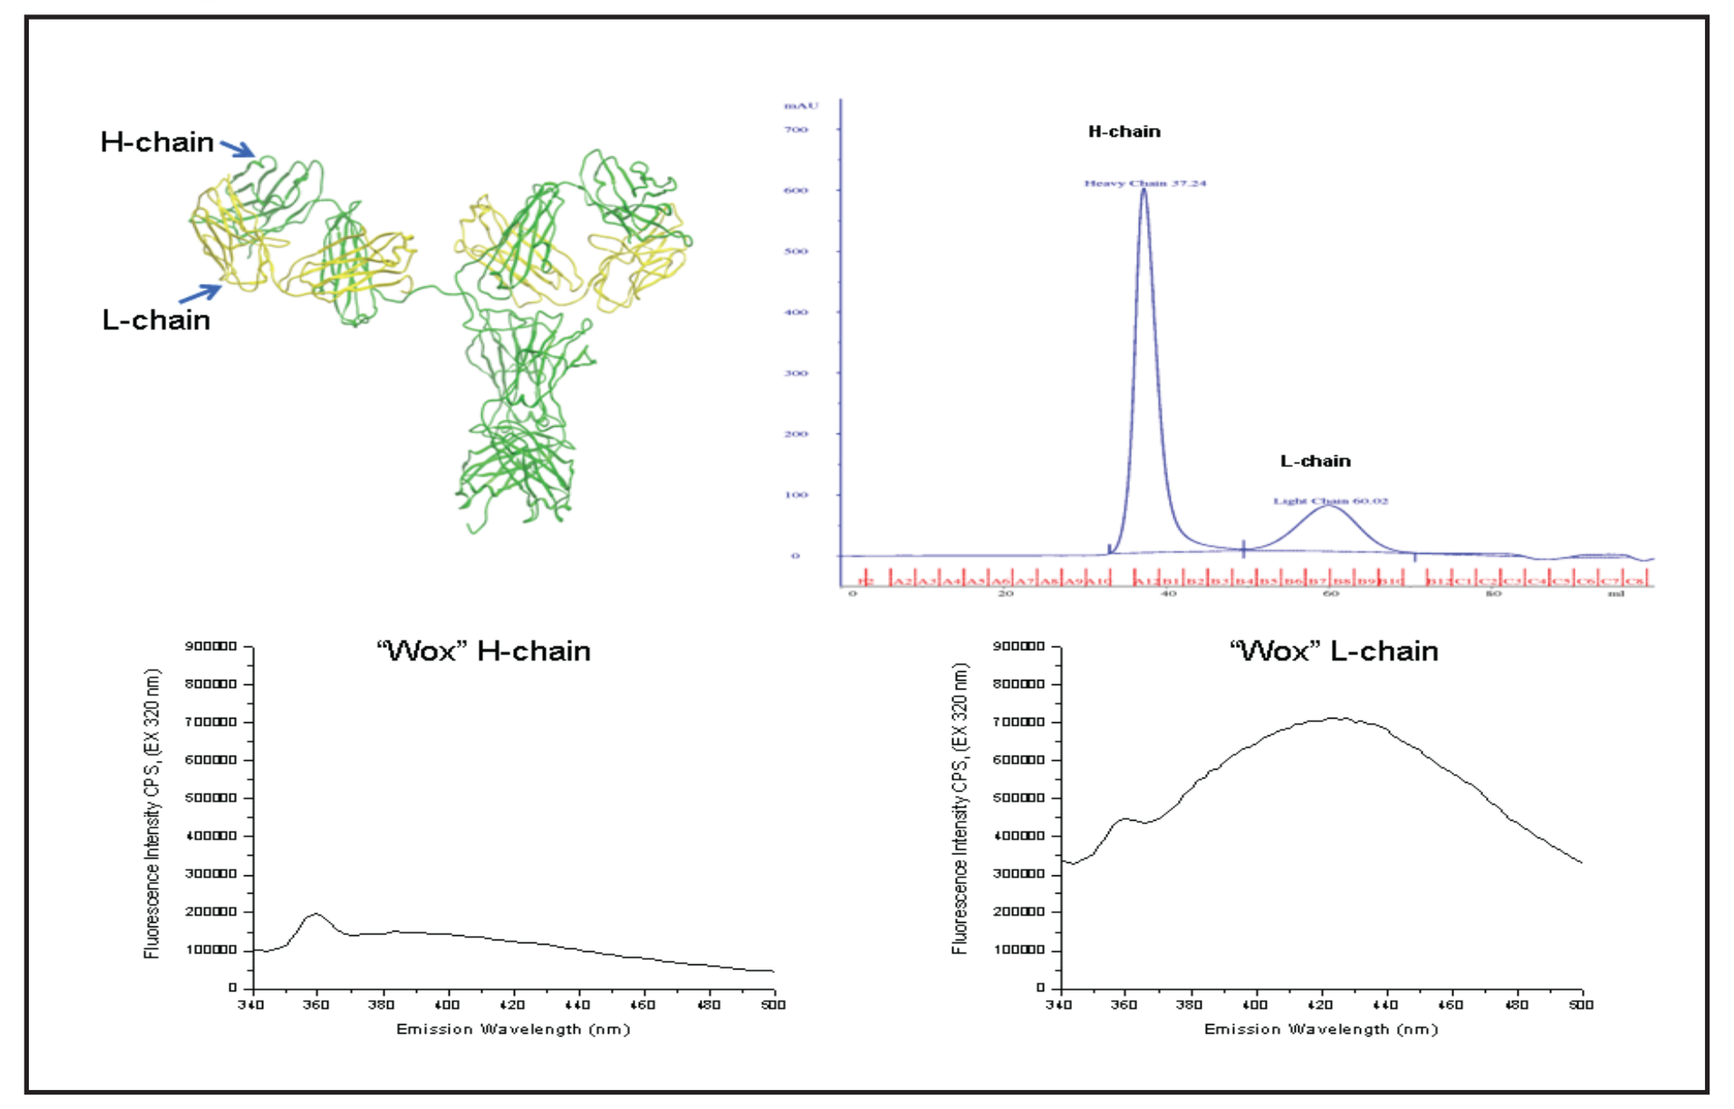

H-chain
L-chain
"Wox" H-chain
"Wox" L-chain
Emission Wavelength (nm)

Upper panels: Heavy and light chain fragments were separated by FPLC size-exclusion chromatography.
Lower panels: Fluorescence emission spectra of "Wox" H-chain and L-chain. Oxidation occurs in L-chain only.

Fig. 7. Simulated structural perturbation following oxidation of individual Trp in light chain fragment

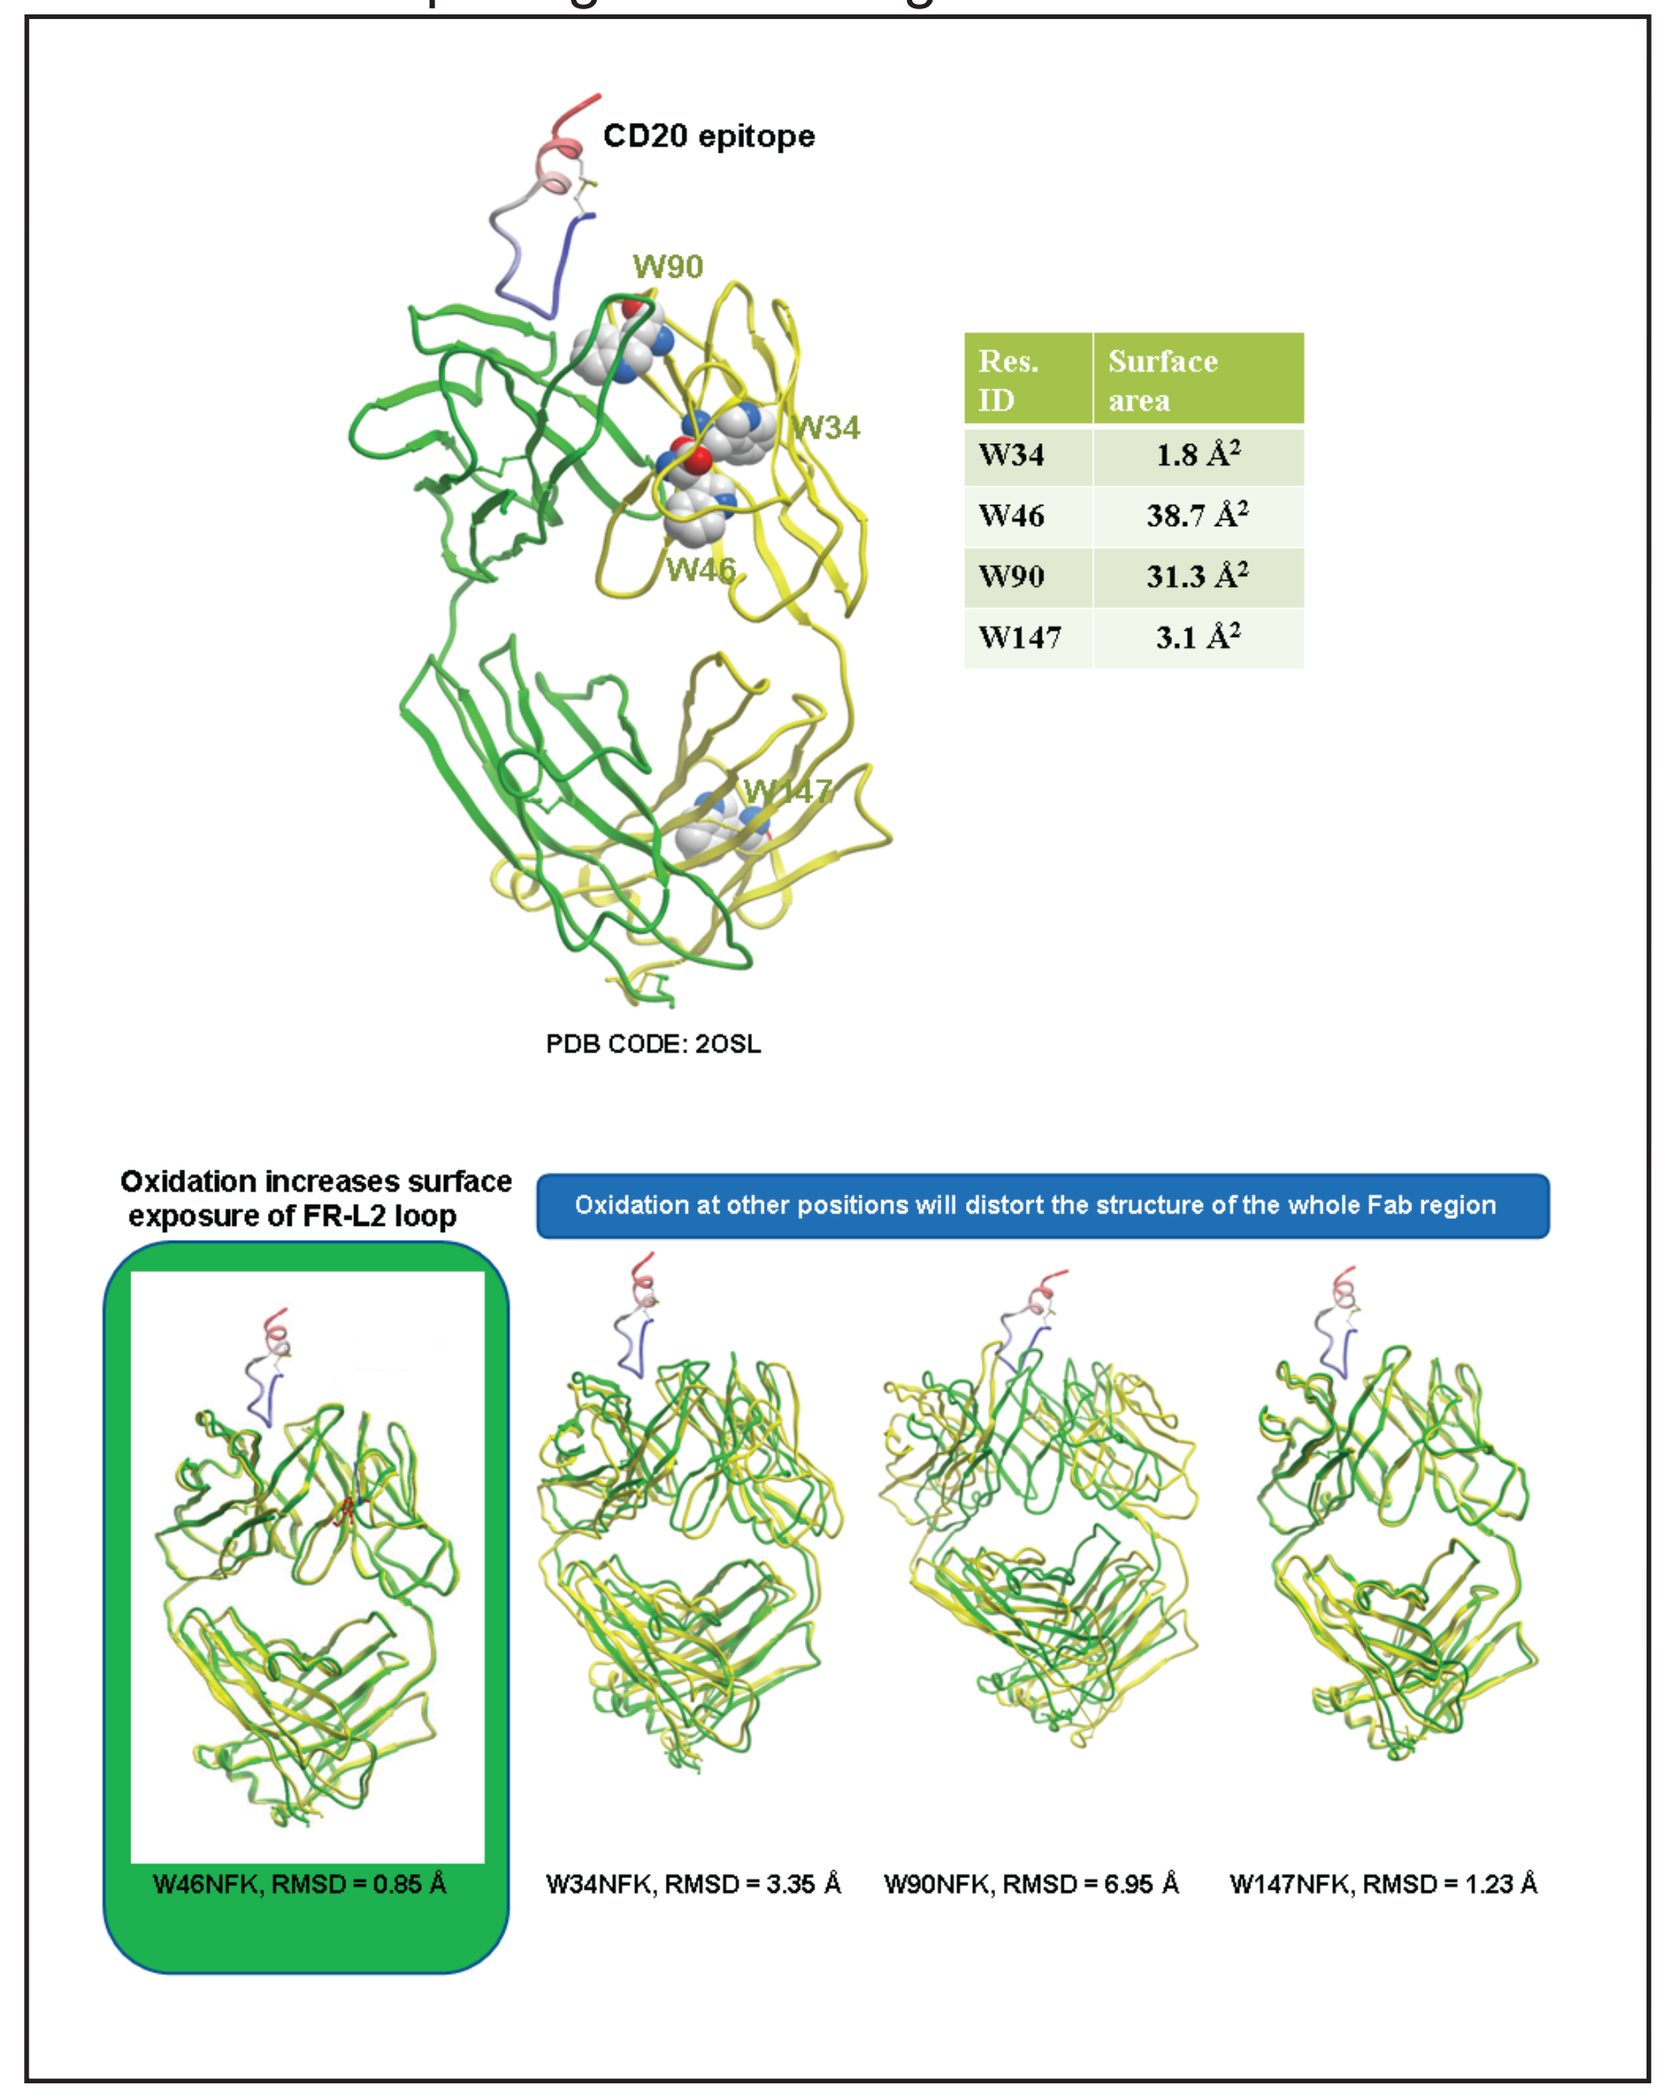

CD20 epitope
W90
W34
W46
W147
PDB CODE: 2OSL
Oxidation increases surface exposure of FR-L2 loop
Oxidation at other positions will distort the structure of the whole Fab region
W46NFK, RMSD = 0.85 Å
W34NFK, RMSD = 3.35 Å
W90NFK, RMSD = 6.95 Å
W147NFK, RMSD = 1.23 Å

Res. ID	Surface area
W34	1.8 Å^2
W46	38.7 Å^2
W90	31.3 Å^2
W147	3.1 Å^2

Upper panels: Calculated surface area of light chain Trps.
Lower panels: Structural perturbation simulated using Monte-Carlo protocol. W46 is most likely to be oxidized based on simulation.

Fig 8. Calculated change of surface area after oxidation of W46

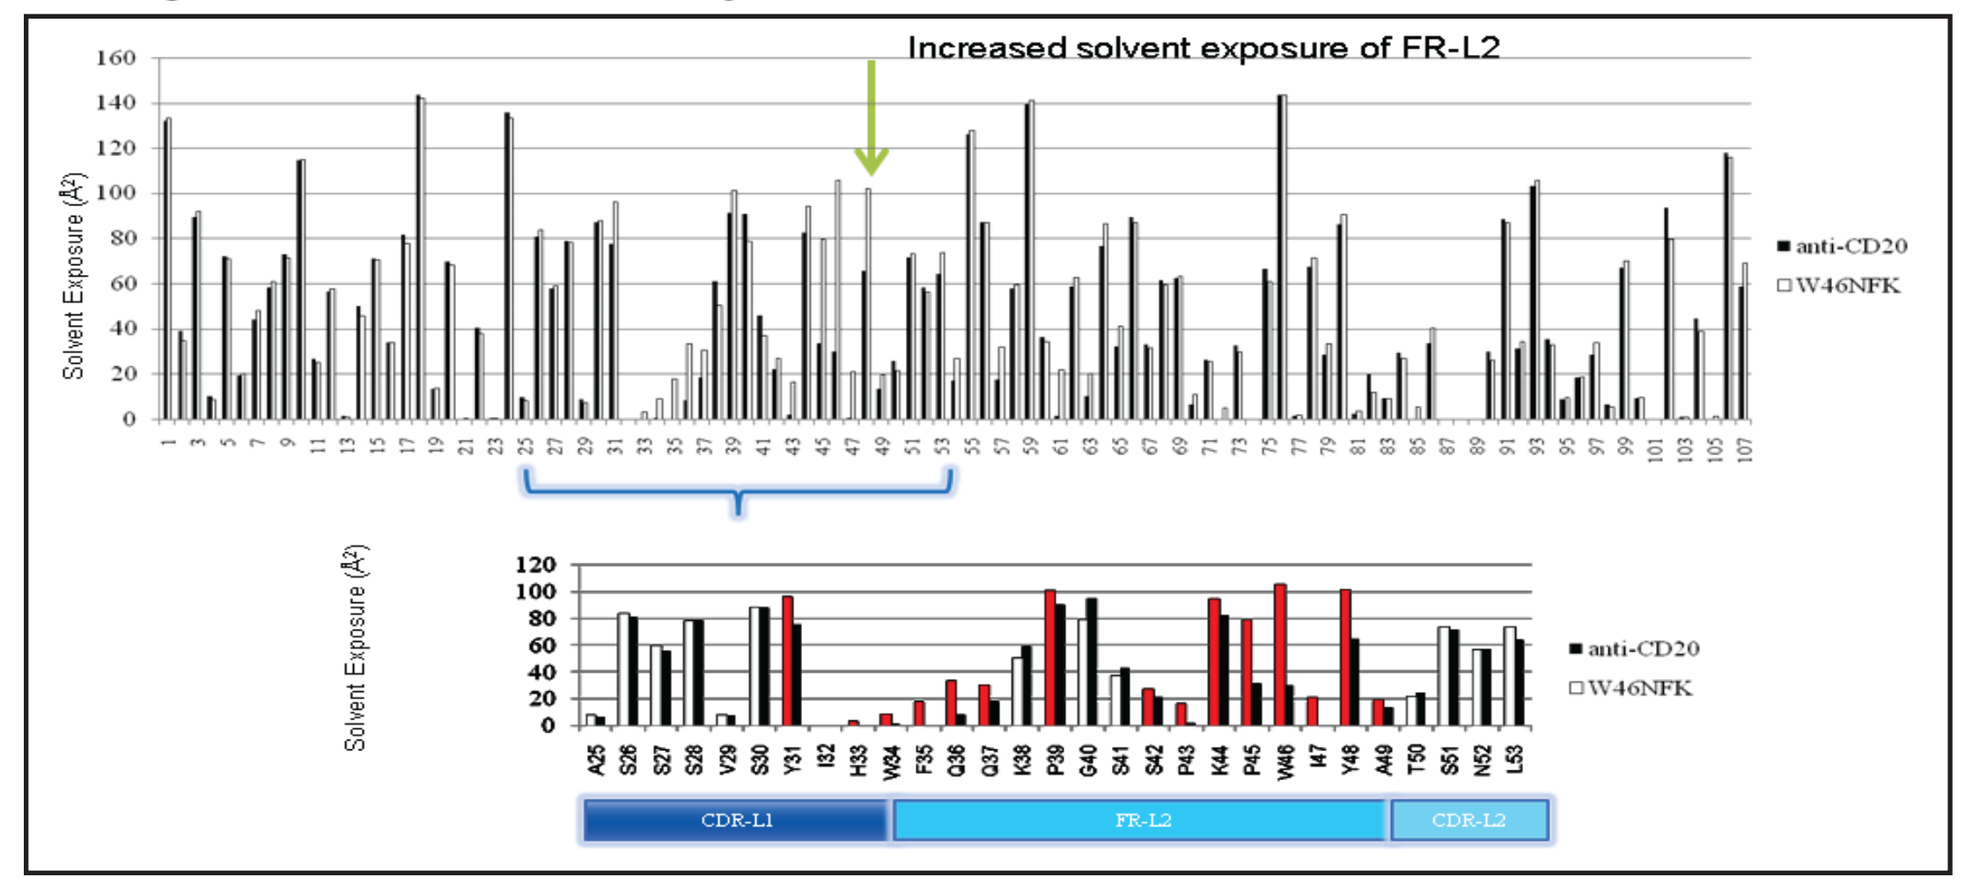

Increased solvent exposure of FR-L2
Solvent Exposure (Å²)
anti-CD20
W46NFK
CDR-L1
FR-L2
CDR-L2

Significant increase of solvent exposure of a variable light chain region between CDRL1-FRL2 (~9% increase for hydrophobic residues in Fab domain).

Conclusions:

1. Oxidation of monoclonal antibodies can be performed in a controlled manner, yielding species (Wox) with improved kinetic profiles without loss of immunogenic specificity.
2. Wox variant antibodies when maintained at 4 ˚C remain stable for at least 6 months.
3. Immunogenicity studies to date (mouse, rabbit, and primate) have shown no formation of anti-drug antibodies specific to the Wox species, suggesting that the modification does not result in the creation of immunogenic neodeterminants.
4. The Wox modification of anti-CD20 antibodies imparts improved potency.

Acknowledgements:

Production & Characterization Group, InNexus Biotechnology, Inc.
Martin Kline, Joe Zdanowicz, Andrew Lassen, Sam Chong,
Marc Bingaman, Tiana Golding